A consistent GABAergic neuron model by optimised reprogramming of human iPSCs

Tony Oosterveen, Dumitra Bors, Muhammad Kaiser Abdul Karim, Daniel Pearce, Hanna Luniak, Tonya Frolov, Farah Patell-Socha, Matthew Hancock, Grant Belgard, Karl Firth, Thomas Moreau, Mark Kotter

bit.bio, The Dorothy Hodgkin Building, Babraham Research Campus, Cambridge, CB22 3FH, UK Corresponding author's e-mail: tony.oosterveen@bit.bio

bit.bio
THE CELL CODING COMPANY

Abstract

Neuronal circuits in the cortex consist of two main types of neurons, namely the glutamatergic principle excitatory cells and the GABAergic inhibitory neurons (INs). The inputs of INs provide cortical networks with the ability to balance spontaneous and evoked excitatory activities, and prevent runaway excitation. Abnormal IN function has been associated with a variety of neurological diseases, including schizophrenia, autism and Alzheimer's disease. For neuronal indications, fewer than 10% of findings derived from animal models can be translated to the clinic[1]. Scalable approaches are needed to generate human in vitro models suitable for high content drug screening that consist of well-defined and pure populations of specific neurons, such as GABAergic neurons. We used our proprietary gene-expression technology, opti-ox[2], to tightly control the expression of ASCL1 and DLX2 and generate a pure population of GABAergic neurons from human iPSCs, at scale, within 14 days. A deep molecular characterisation of these neurons by immunocytochemistry, RT-qPCR and single cell RNA-sequencing revealed that the cultures consist of over 95% of GABAergic neurons expressing the classical marker genes GAD1, GAD2, VGAT, DLX1 as well as DLX2 and are positive for GABA. Functional annotation of genes enriched in the post-mitotic GABAergic neurons identified gene-sets significantly associated with several neurological disorders linked to INs, including schizophrenia, autism and ADHD. Anchoring our single cell RNA-seq data of the iPSC-derived GABAergic neurons on the Allen Brain single cell map of the human cortex showed that these neurons closely match with the VIP subtype of GABAergic neurons. A functional analysis by MEA demonstrated that they form neuronal networks and spontaneously fire action potentials within 2 to 3 weeks. Our highly consistent and synchronised opti-ox cellular reprogramming enables us to manufacture highly pure GABAergic neurons in large quantities in a reproducible manner. opti-ox reprogramming technology is being applied to a range of other neuronal subtypes and opens up novel avenues for the development of more reliable human in vitro models to support research and healthcare innovations.

1. Precise reprogramming of iPSCs into defined human cell types

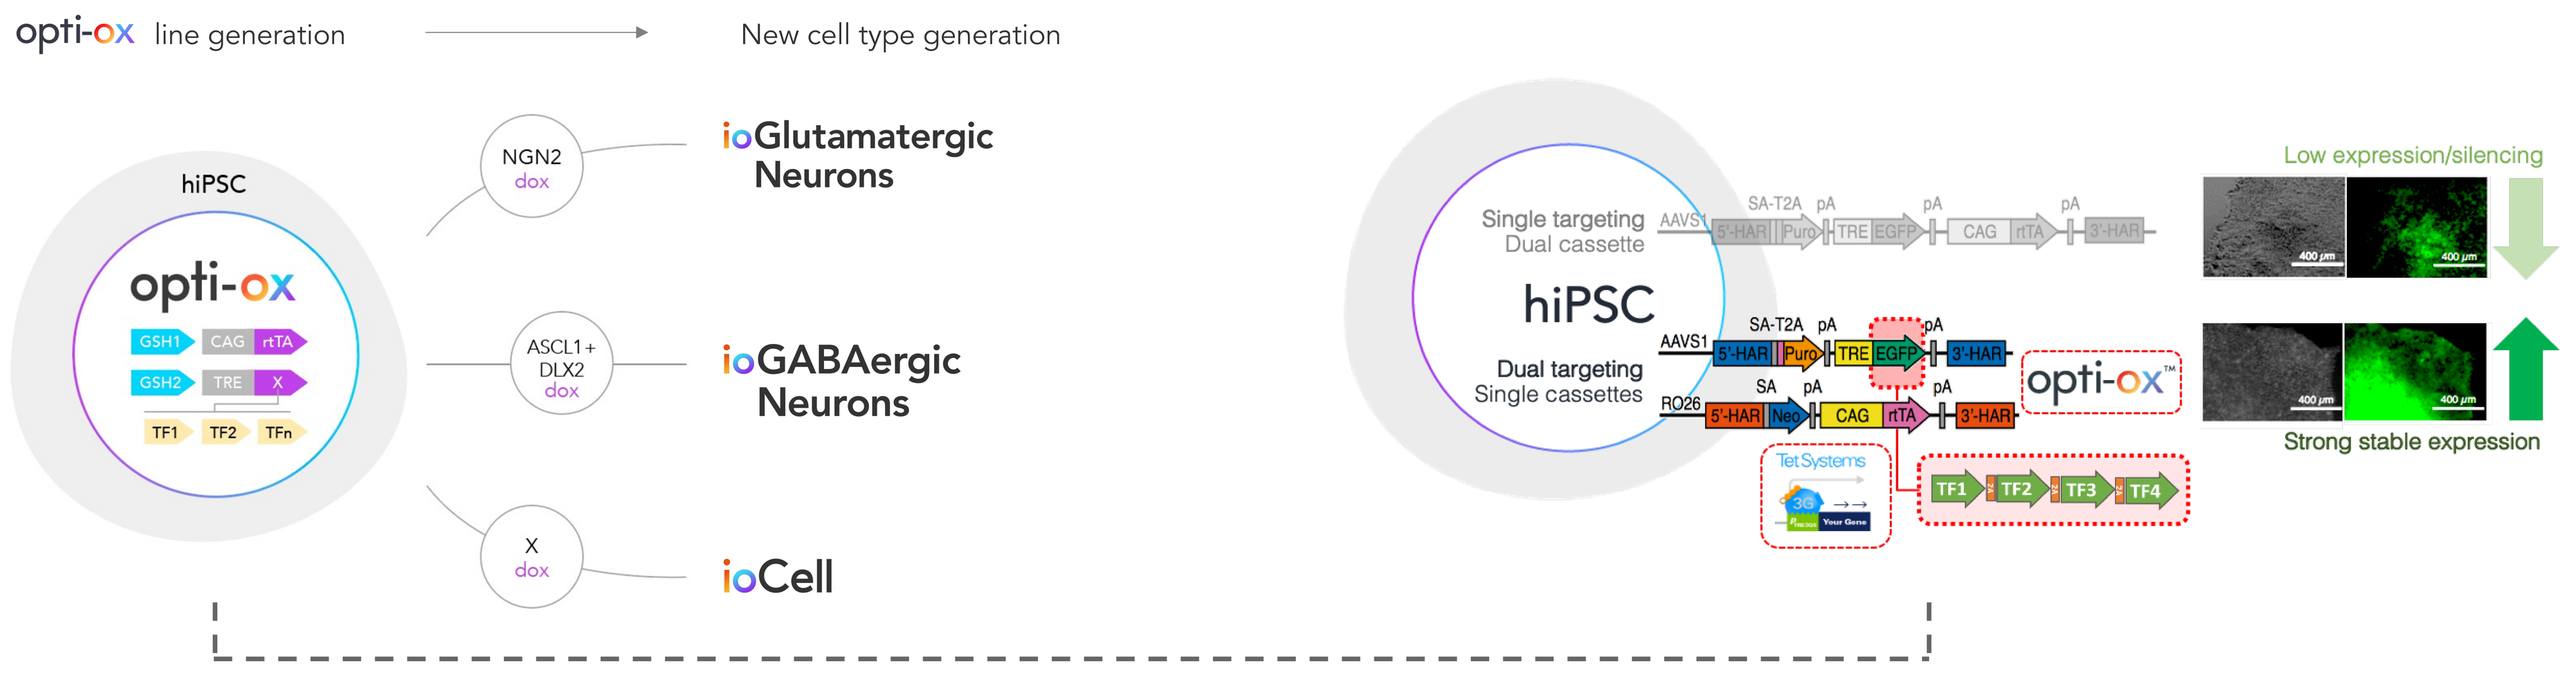
opti-ox line generation
New cell type generation
hiPSC
opti-ox
NGN2 dox
ioGlutamatergic Neurons
ASCL1+ DLX2 dox
ioGABAergic Neurons
X dox
ioCell
Single targeting Dual cassette
hiPSC
Dual targeting Single cassettes
Low expression/silencing
Strong stable expression

Figure 1. opti-ox technology for the optimal cellular reprogramming of human iPSCs into defined human cell types. opti-ox dual cassette Tet- ON system ensures tightly controlled and homogeneous expression of reprogramming transcription factors by preventing silencing of the inducible expression cassette after genetic engineering of hiPSCs[2].

2. Ready to culture human GABAergic Neurons ready for experimentation within days

Figure 2. ioGABAergic Neurons are derived from hiPSCs by ASCL1 and DLX2 driven opti-ox reprogramming and arrive ready to plate. **(A)** Cells are delivered in a cryopreserved format and are programmed to rapidly mature upon revival in the recommended media. The protocol for the generation of these cells is a three-phase process: 1. Induction (carried out at bit.bio); 2. Stabilization for 3 days with Doxycycline; 3. Maintenance during which the GABAergic neurons mature. **(B)** Images show rapid morphological changes in the cells upon reprogramming, with neurons identified by Day 7 post-revival. Mature neuronal networks are observed by Day 11. 10X magnification; scale bar: 200µm.

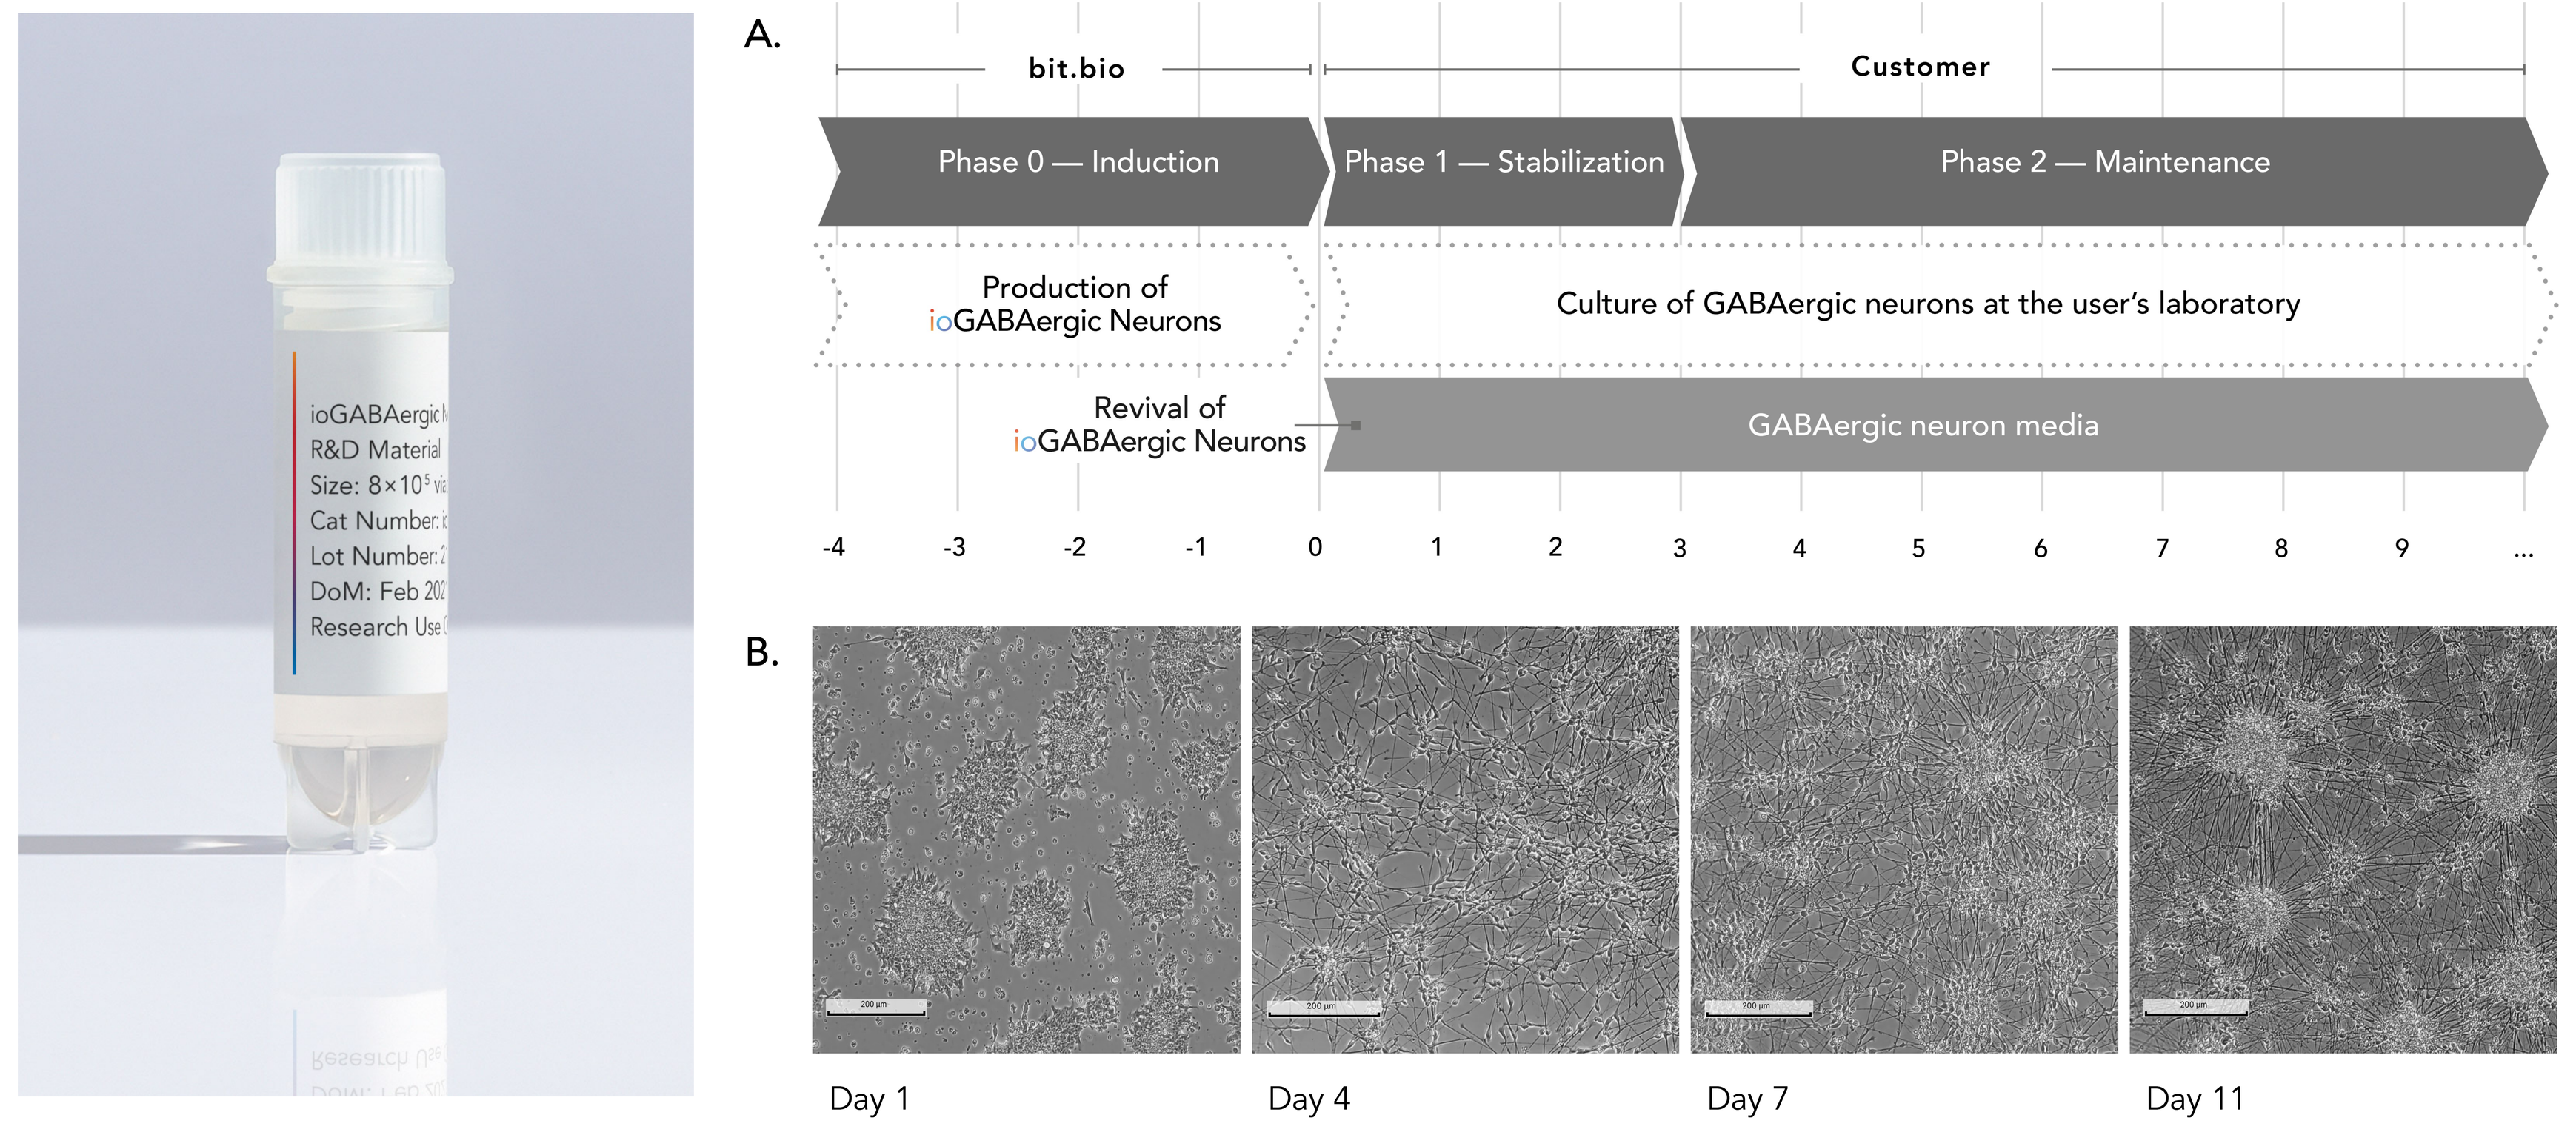
A.
bit.bio
Customer
Phase 0 — Induction
Phase 1 — Stabilization
Phase 2 — Maintenance
Production of ioGABAergic Neurons
Culture of GABAergic neurons at the user's laboratory
Revival of ioGABAergic Neurons
GABAergic neuron media
-4 -3 -2 -1 0 1 2 3 4 5 6 7 8 9 ...
B.
Day 1
Day 4
Day 7
Day 11

3. Mature properties within one week of induction

Figure 3: Characterization of ioGABAergic Neurons. Antibodies against key marker molecules specific for GABAergic neurons show that ioGABAergic neurons are positive for GABA and VGAT1 as well as for the general neuronal marker MAP2 after 7days.

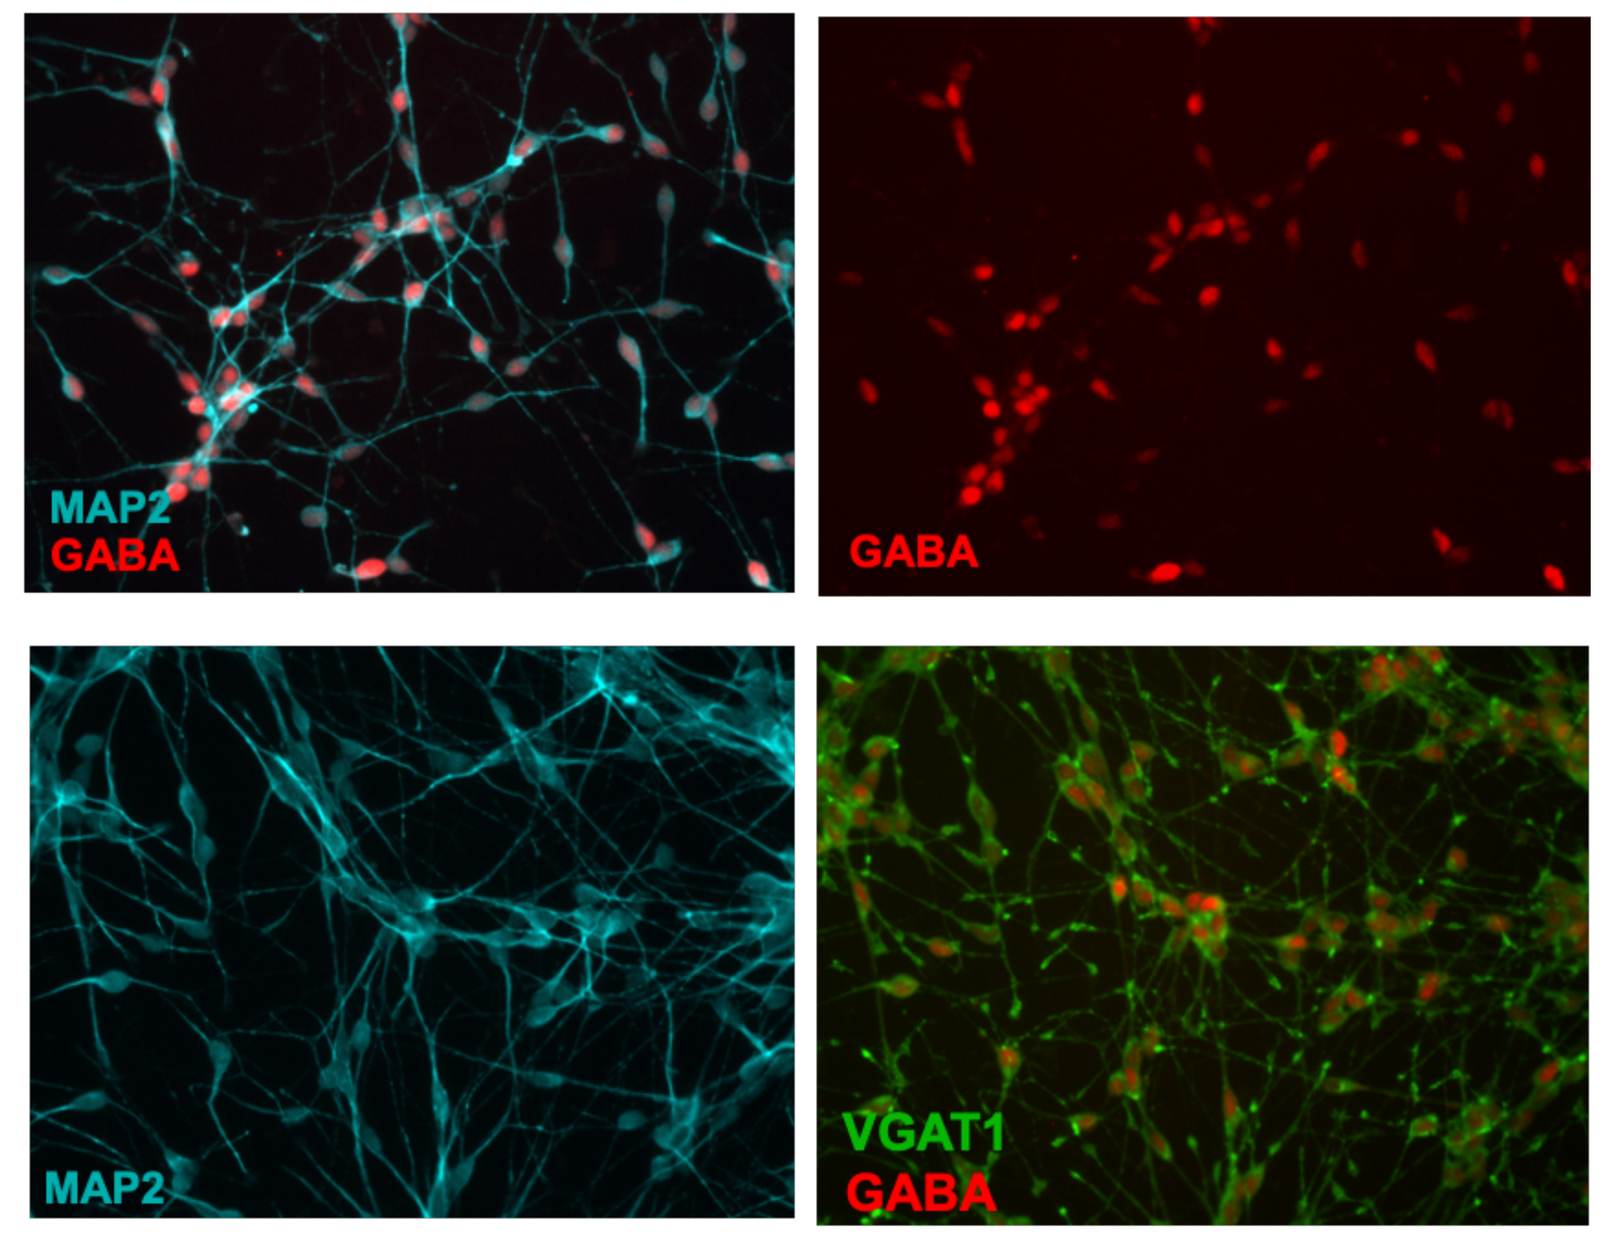
MAP2
GABA
GABA
MAP2
VGAT1
GABA

4. Expression of ASCL1 and DLX2 efficiently convert iPSCs into GABAergic neurons

Figure 4: Characterisation of the ioGABAergic Neurons at the single cell level by RNA-sequencing. Analysis of different time-points during the reprogramming of human iPSCs into GABAergic neurons. The UMAP plot clusters cells based on progressive acquisition of the GABAergic neuron phenotype. The day 14 cluster in red contains only post-mitotic neurons and shows a high purity of GABAergic neurons (>95%).

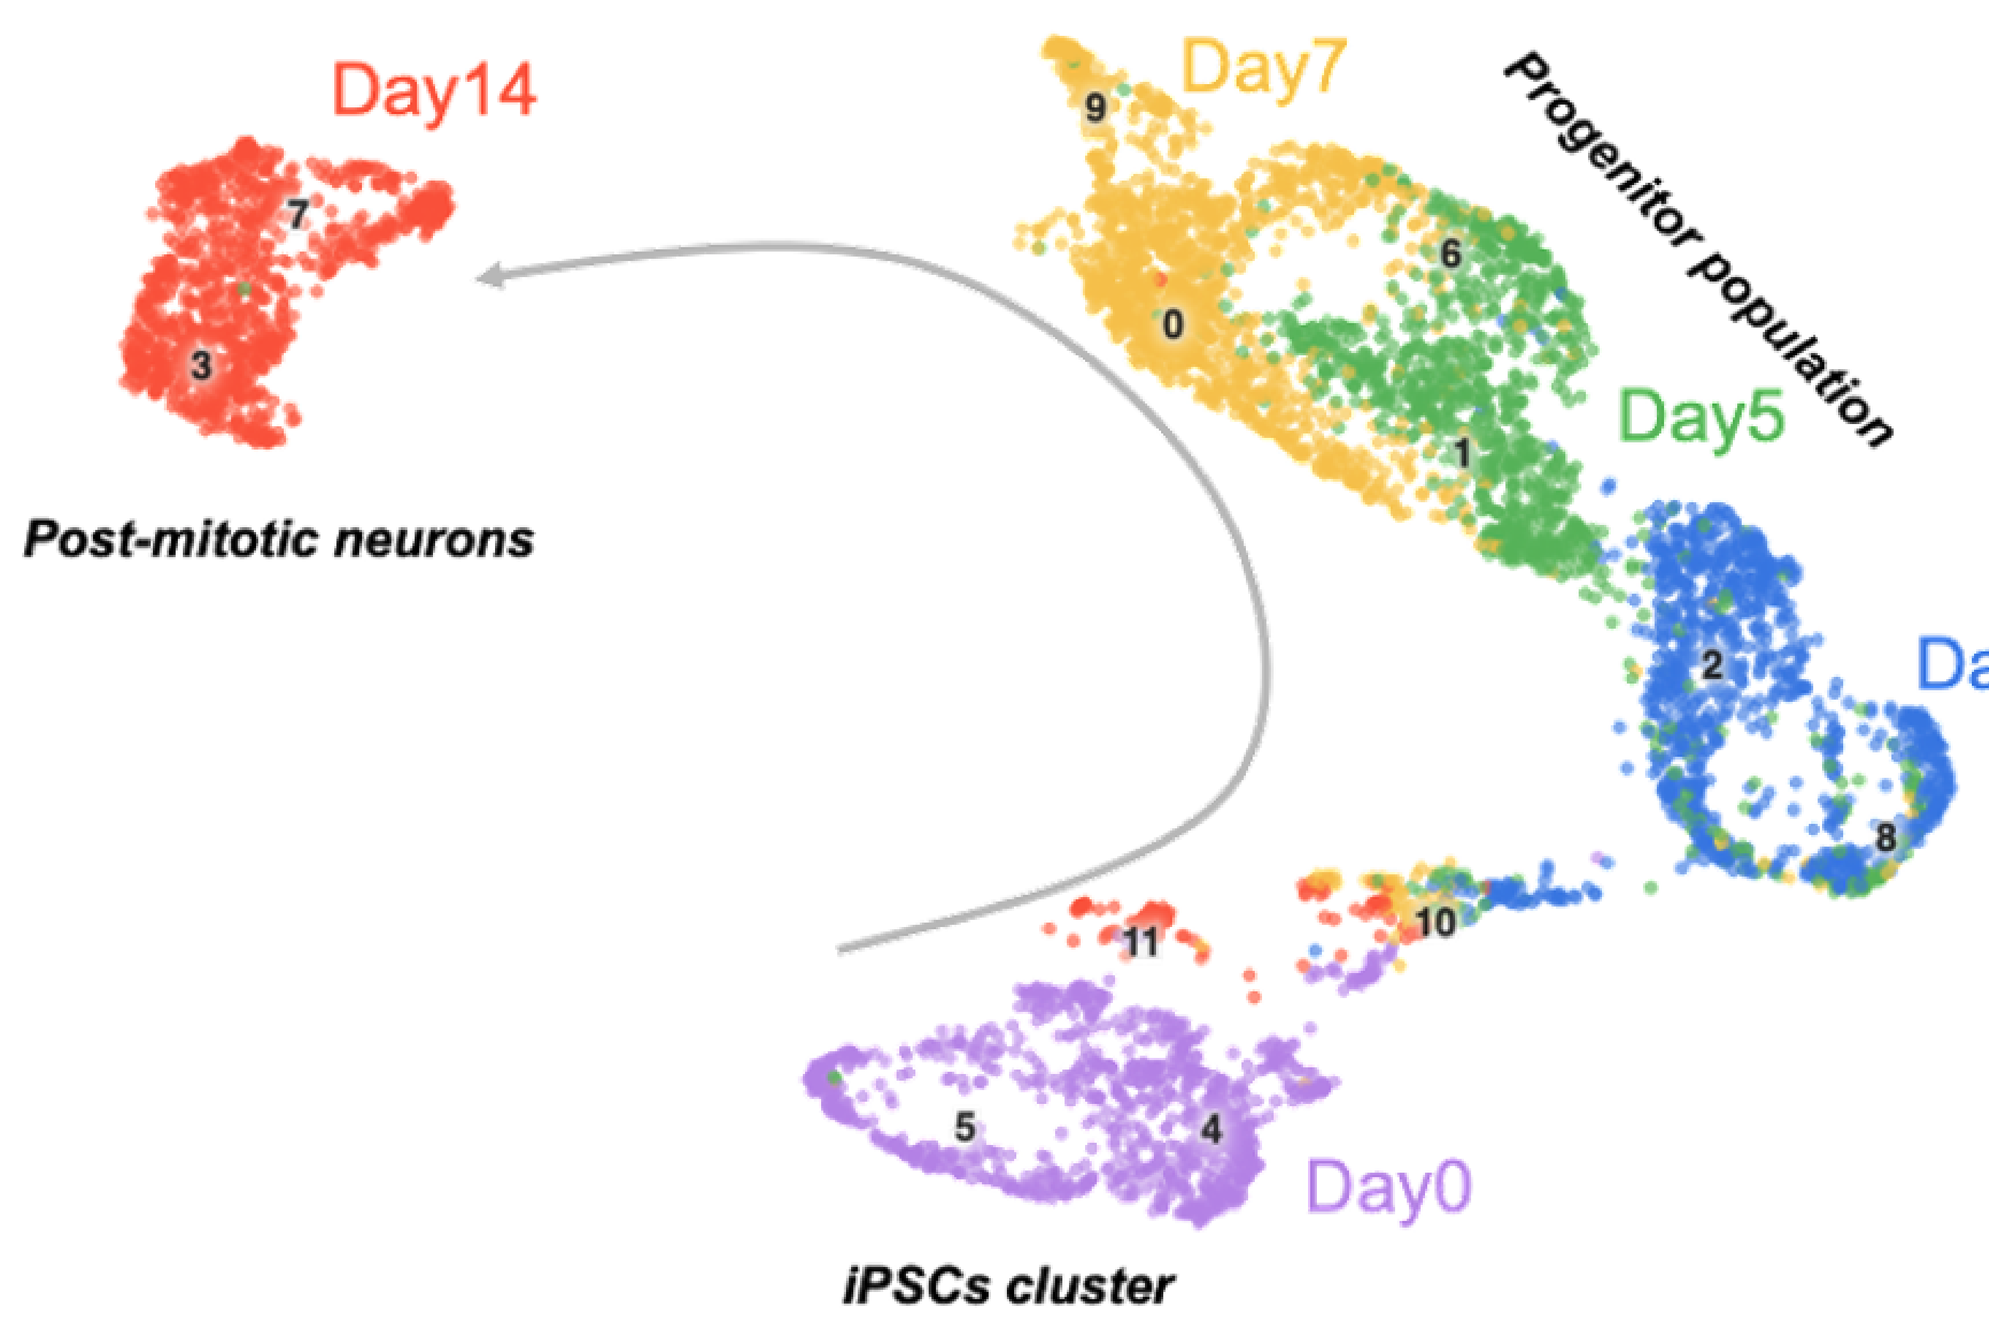
Day14
Post-mitotic neurons
Day7
Progenitor population
Day5
Day3
Day0
iPSCs cluster

5. Highly-defined and characterized by single cell RNA sequencing

Figure 5: Expression profile of key marker genes for GABAergic neurons detected by single cell RNAseq. The left panel shows that the day 14 post-mitotic neurons express key marker genes of GABAergic neurons (DLX1, GAD1, GAD2, VGAT) together with pan-neuronal marker MAP2. On the right panel, the dataset demonstrates that markers of other neuronal populations (Glutamatergic, Dopaminergic, Serotonergic and adrenergic neurons) are absent or barely expressed.

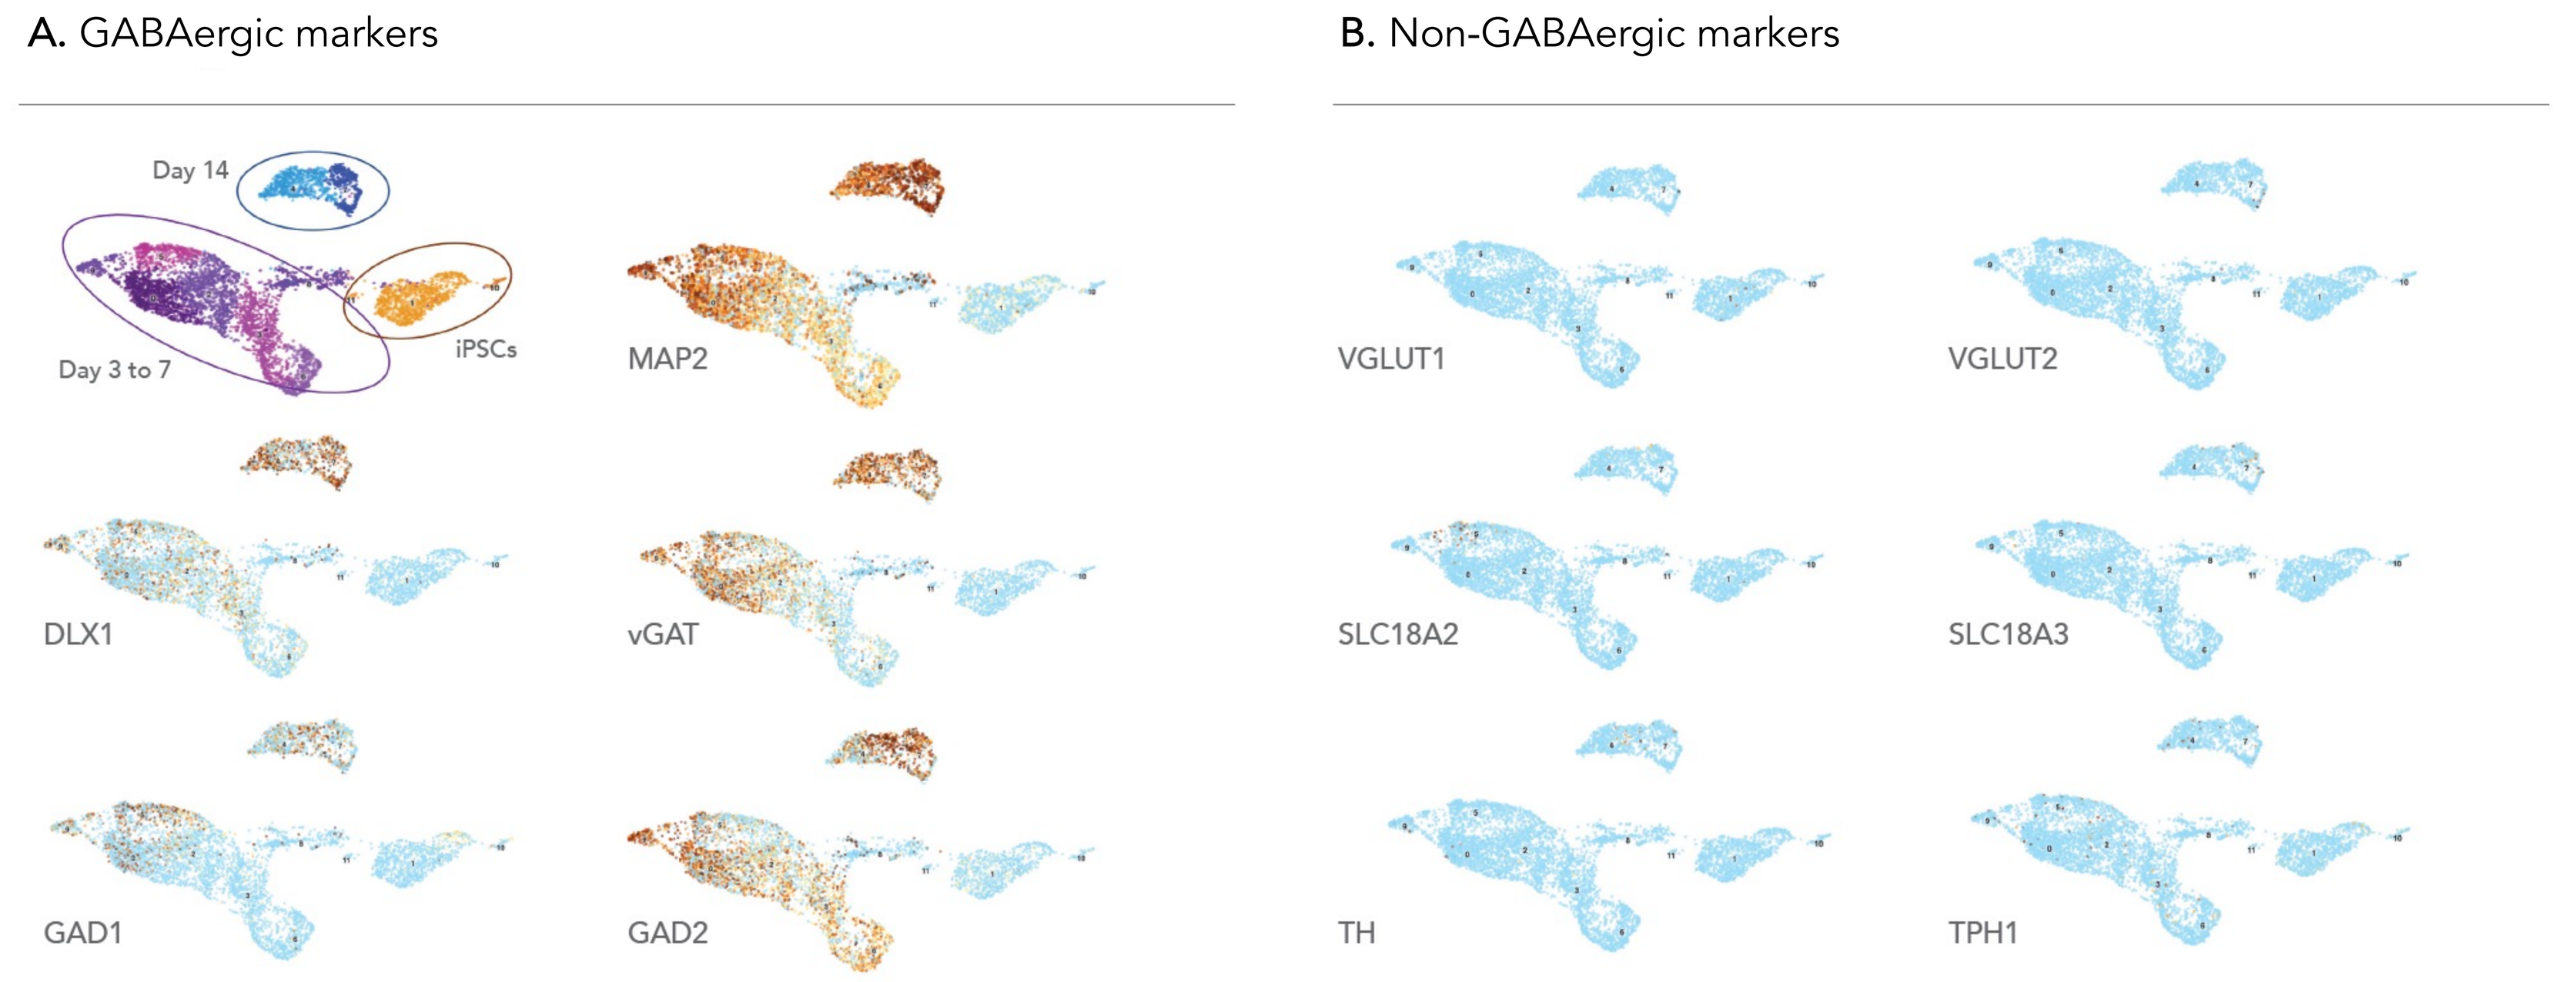
A. GABAergic markers
B. Non-GABAergic markers
Day 14
Day 3 to 7
iPSCs
MAP2
DLX1
vGAT
GAD1
GAD2
VGLUT1
VGLUT2
SLC18A2
SLC18A3
TH
TPH1

6. Bioinformatic analysis of GABAergic subtype identity reveals cells belong to the VIP subtype

Figure 6: Bioinformatic analysis of GABAergic subtype identity from single cell RNA-seq data. Anchoring our single cell RNA-seq data of the iPSC-derived GABAergic neurons on the Allen Brain single cell map of the human cortex demonstrates that the ioGABAergic Neurons (red cluster) closely match with the VIP subtype of GABAergic neurons (purple cluster).

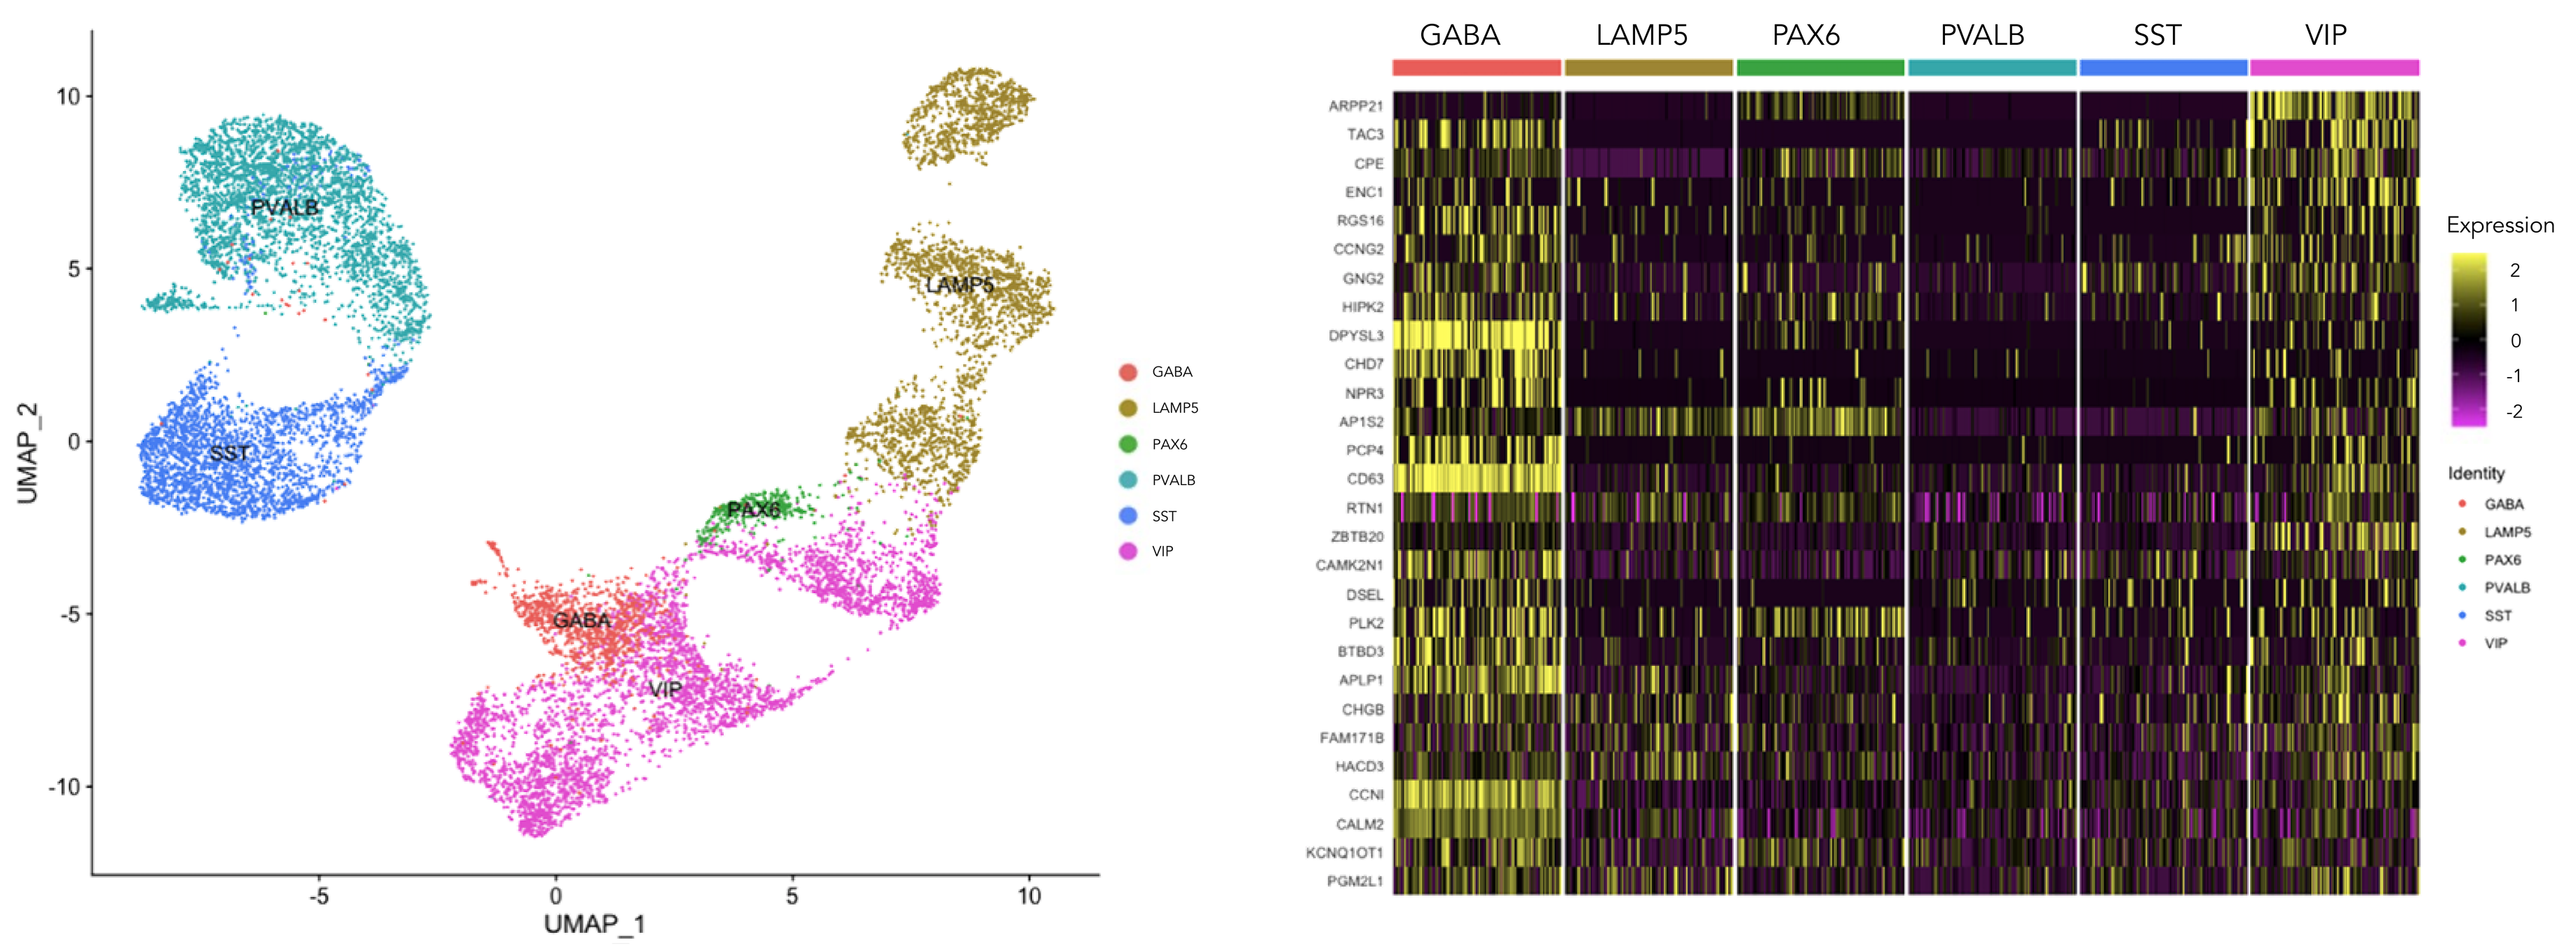
UMAP_2
UMAP_1
PVALB
SST
LAMP5
PAX6
GABA
VIP
GABA
LAMP5
PAX6
PVALB
SST
VIP
Expression
Identity

7. Spontaneous neuronal activity is detected within 20 days post-revival

Figure 7: Functional activity of the ioGABAergic Neurons demonstrated by MEA. 3Brain high-resolution multi electrode array (MEA) recordings of cocultures of rat astrocytes and ioGABAergic Neurons at day 20 post-revival. The snapshots are taken from a 10-minute recording, each snapshot shows on the left a heatmap of the active electrodes on the chip and on the right 3 traces of active electrode and one of an inactive electrode. These data show that ioGABAergic Neurons fire spontaneously and can form functional neuronal networks

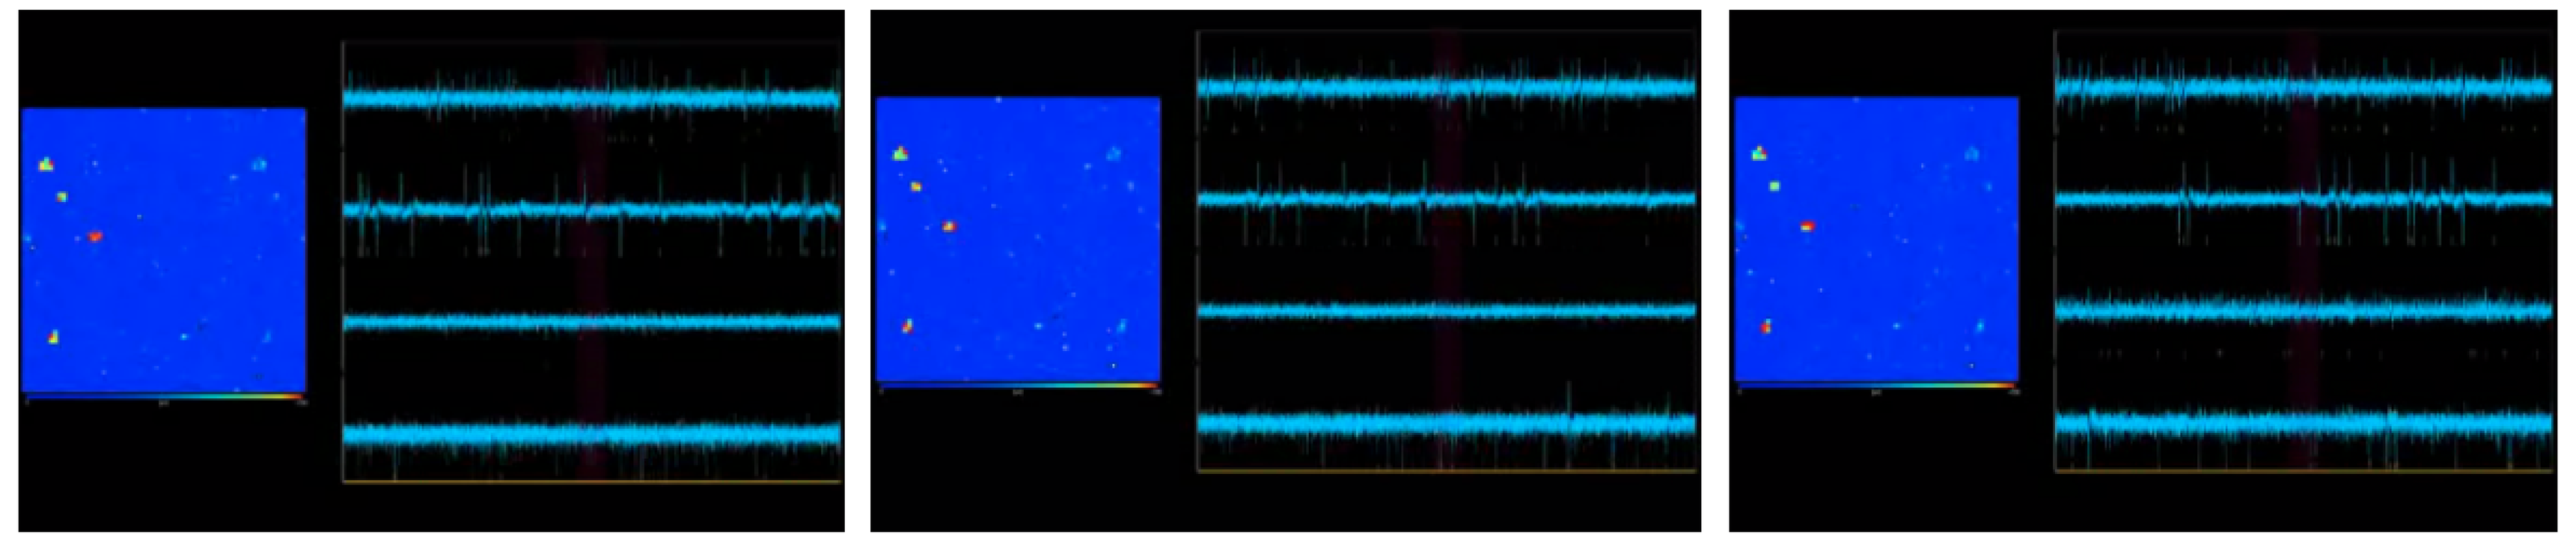

1. Vargas-Caballero M, et al., Expert Opin Drug Discov. 2016;11(4):355-67. **2.** Pawlowski M, et al., Stem Cell Reports, 2017.

bit.bio I The Dorothy Hodgkin Building I Babraham Research Campus I Cambridge I CB22 3FH I UK I info@bit.bio